Assessment of Pharmacokinetics and Safety of Investigational Nitric Oxide-Releasing SB204 Gel in Adolescents With Acne Vulgaris

Diane Thiboutot[1], Andrea Zaenglein[2], Adelaide Hebert[3], Lawrence Eichenfield[4]

[1]Department of Dermatology, Penn State Hershey Medical Center, Hershey, PA, [2]Department of Dermatology and Pediatrics, Penn State Hershey Medical Center, Hershey, PA, [3]Department of Dermatology, The University of Texas Medical School, Houston, TX, [4]Department of Dermatology, University of California, San Diego, CA

Introduction

- SB204, a nitric oxide releasing topical drug candidate, is in development for the treatment of acne vulgaris
- The active ingredient in SB204 is NVN1000, a polysiloxane macromolecule that stores nitric oxide on the polymer backbone
- SB204 has potential immunomodulating and broad-spectrum antimicrobial activity
- Three studies with SB204 4% applied **once daily** in adolescents assessed:
 - Systemic exposure (NI-AC103) to
 - hydrolyzed N-methyl-aminopropyltrimethoxysilane (hMAP3), a component of NVN1000
 - Nitrate, a marker for nitric oxide
 - Safety (NI-AC103, NI-AC301 and NI-AC302)
 - Efficacy (NI-AC301 and NI-AC302)

Immunomodulatory and Antimicrobial Activity of Nitric Oxide in Acne

- Nitric oxide inhibits the NLRP3 inflammasome, decreasing the downstream release of IL-1β and IL-17, as well as, kills *P.* acnes

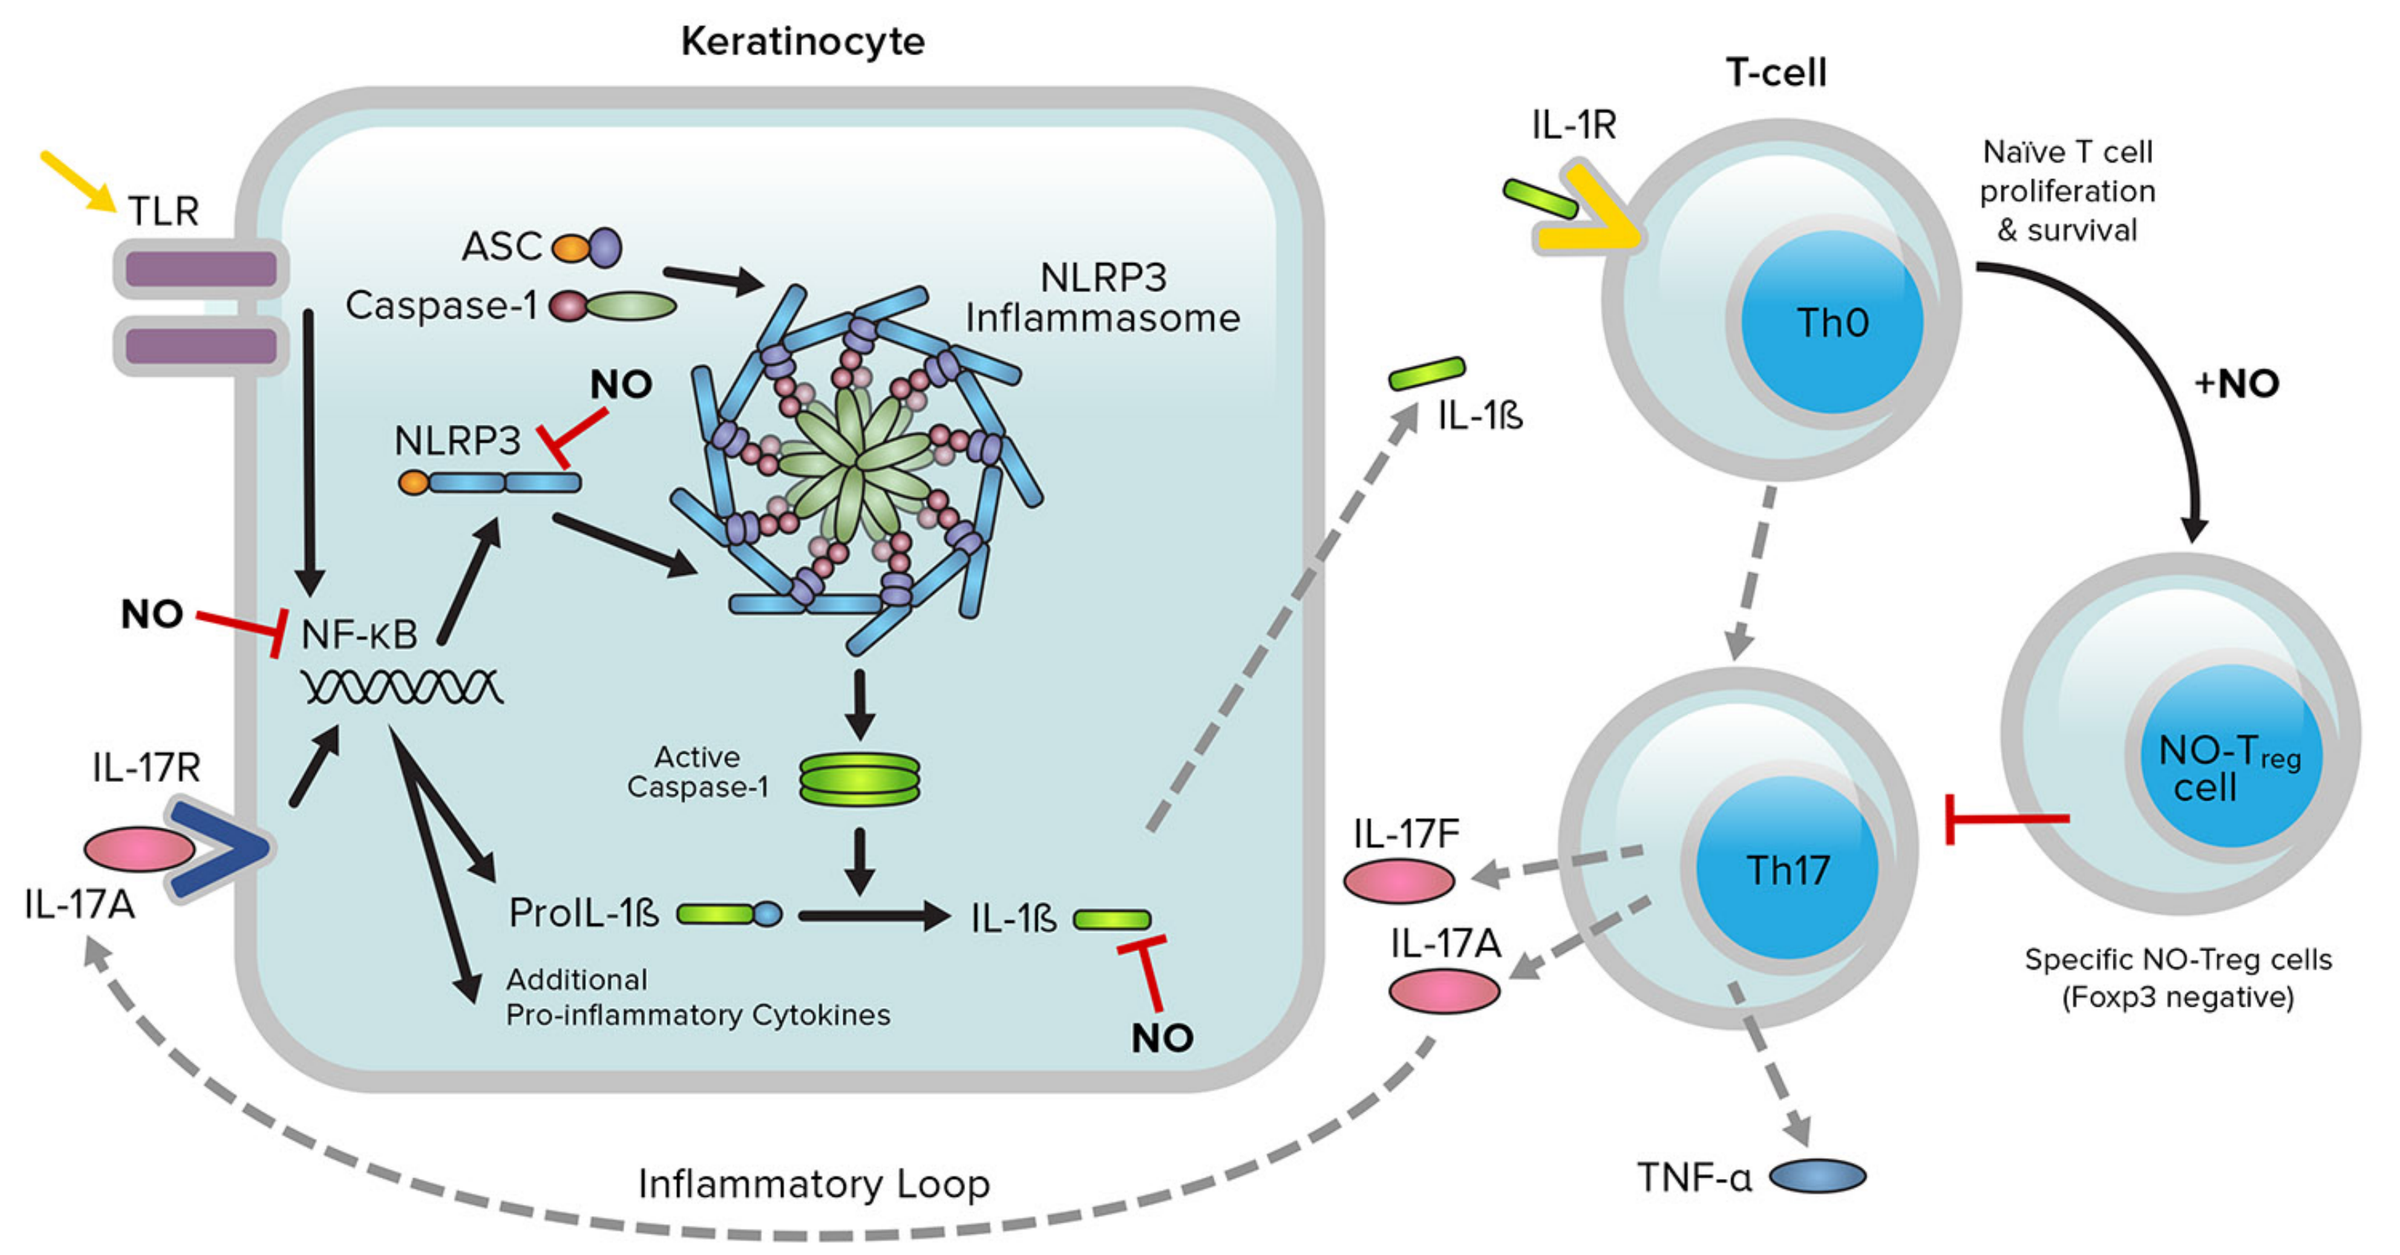
Keratinocyte
TLR
ASC
Caspase-1
NLRP3
Inflammasome
NLRP3
NO
NO
NF-κB
IL-17R
IL-17A
Active
Caspase-1
ProIL-1ß
IL-1ß
Additional
Pro-inflammatory Cytokines
NO
IL-1ß
T-cell
IL-1R
Th0
Naïve T cell
proliferation
& survival
+NO
NO-Treg cell
Specific NO-Treg cells
(Foxp3 negative)
Th17
IL-17F
IL-17A
TNF-α
Inflammatory Loop

Mishra B et al. Nitric oxide controls the immunopathology of tuberculosis by inhibiting NLRP3 inflammasome–dependent processing of IL-1β. *Nature Immunology*. 2013;14:52-60.
Niedbala W et al. Regulation of Type 17 Helper T-Cell Function by Nitric Oxide During Inflammation *Proc Natl Acad Sci USA*. 2011;108(22):9220-9225.
Niedbala W et al. Nitric Oxide-Induced Regulatory T Cells Inhibit Th17 but Not Th1 Cell Differentiation and Function. *J Immunol*. 2013;191(1):164-170.
Qin M et al. Nitric Oxide Releasing Nanoparticles Prevent Propionibacterium Acnes Induced Inflammation by Both Clearing the Organism and Inhibiting Microbial Stimulation of the Innate Immune Response. *J Invest Dermatol*. 2015;135(11):2723-2731.

NI-AC301/302

NI-AC301/302 STUDY OVERVIEW

- Studies in 2600 subjects with moderate to severe acne. Data presented is from subset of 905 adolescents, ages ≥9 - <17
- Subjects were randomized 1:1 ratio of SB204 4% or vehicle

Randomized (N [pooled] = 905)
- SB204 4% gel once daily (n [pooled] = 439)
- Vehicle gel once daily (n [pooled] = 466)

12 weeks

- SB204 4% gel (900mgs) or vehicle (900mgs) were applied once daily to the entire face
- Primary efficacy endpoints assessed:
 - Absolute change in inflammatory and noninflammatory lesion counts from baseline to week 12
 - Success on Investigator's Global Assessment (IGA) at week 12 (IGA success was defined as a score of clear (0) or almost clear (1) and ≥2 grades less than baseline)

NI-AC301/302 Demographics

NI-AC301 and NI-AC302	SB204 4% (n, pooled = 439)	Vehicle once daily (n, pooled = 466)
Gender, n		
Male	228 (52%)	255 (55%)
Female	211 (48%)	211 (45%)
Age, mean	14	14
Baseline, mean (SD)		
Inflammatory Lesion	28 (5.7)	28 (5.9)
Non-Inflammatory Lesion Count	42 (13)	42 (13)
Total Lesions	70 (15)	70 (16)
Baseline IGA Scores		
"Moderate" or a score of 3	377 (86%)	401 (86%)
"Severe" or a score of 4	62 (14%)	65 (14%)
Disposition, n		
Completed	397 (90%)	421 (90%)
Discontinued	42 (10%)	45 (10%)

NI-AC301/302 Results: Tolerability

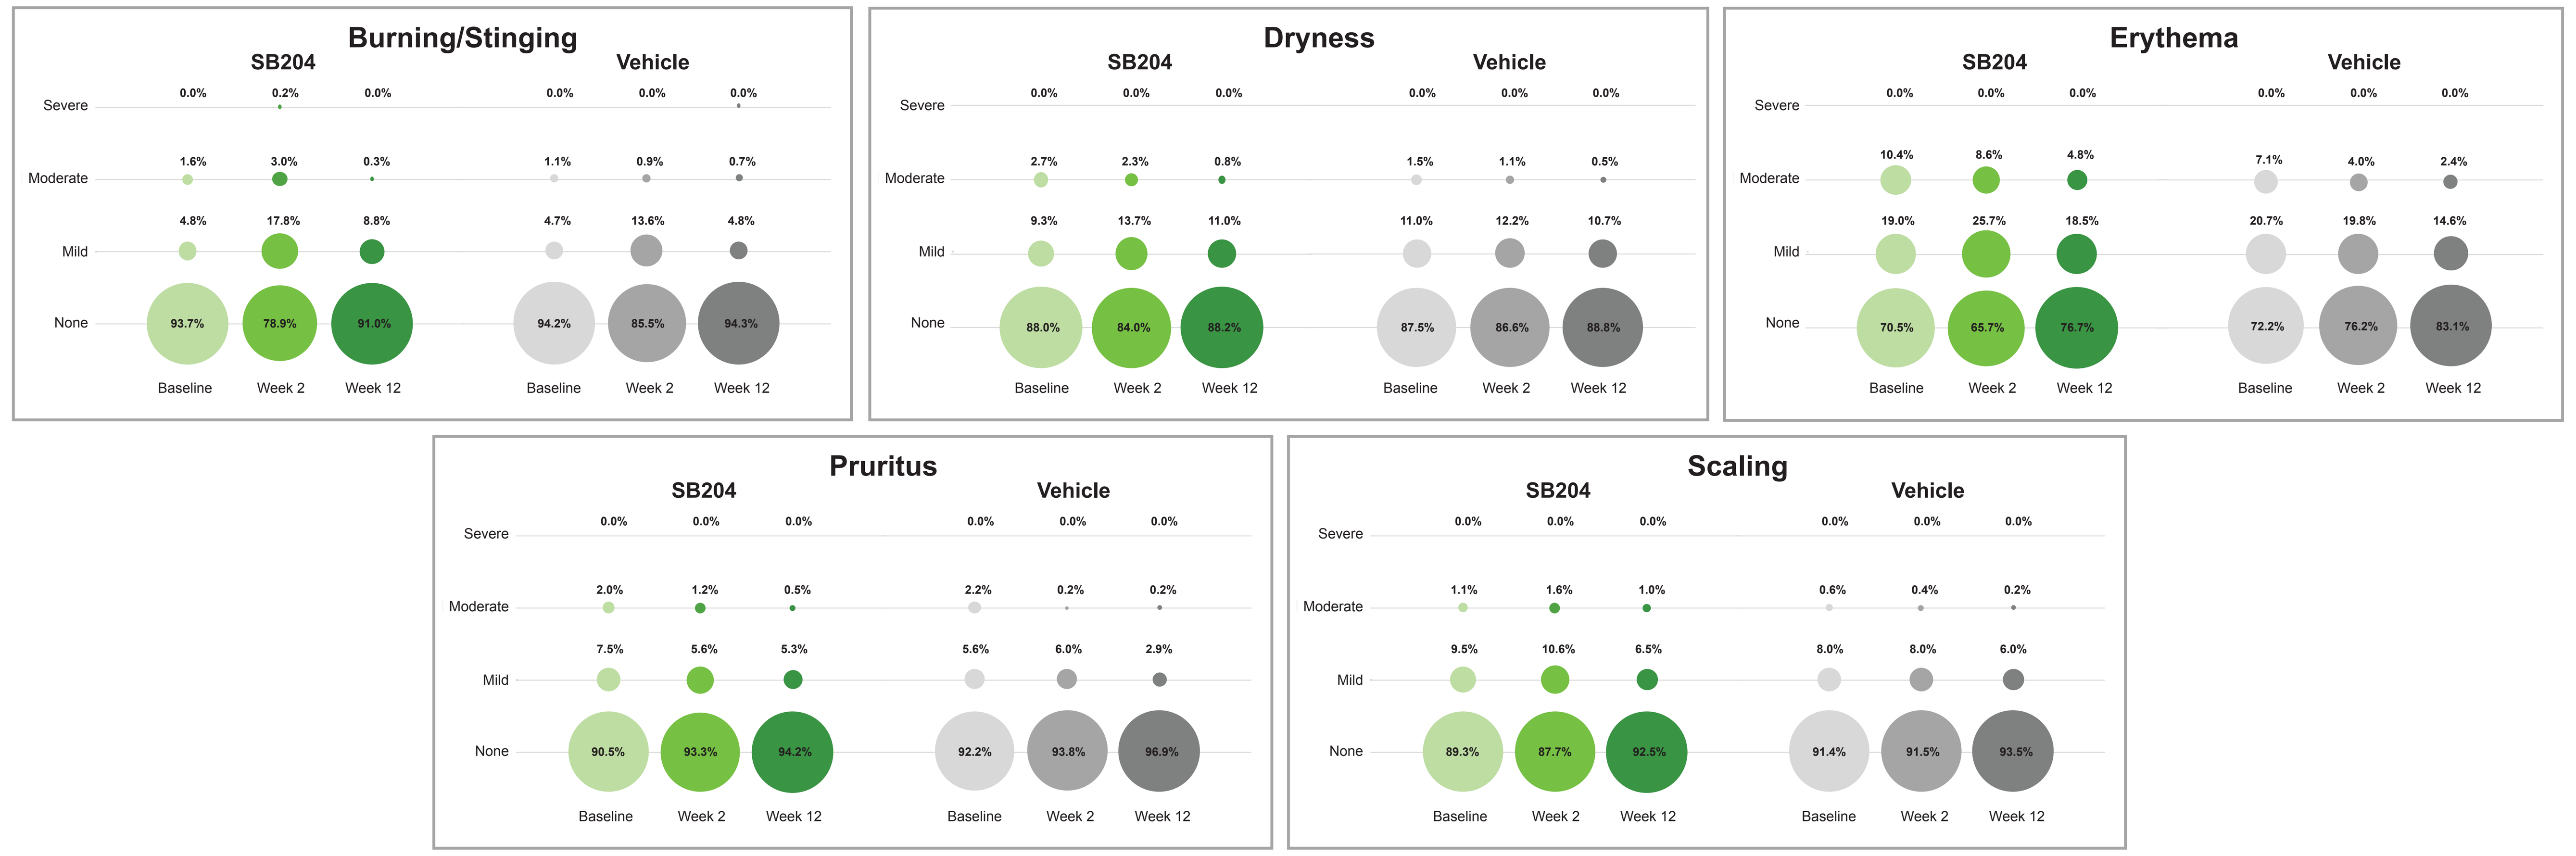
Burning/Stinging
SB204
Vehicle
Severe 0.0% 0.2% 0.0% 0.0% 0.0% 0.0%
Moderate 1.6% 3.0% 0.3% 1.1% 0.9% 0.7%
Mild 4.8% 17.8% 8.8% 4.7% 13.6% 4.8%
None 93.7% 78.9% 91.0% 94.2% 85.5% 94.3%
Baseline Week 2 Week 12 Baseline Week 2 Week 12
Dryness
SB204
Vehicle
Severe 0.0% 0.0% 0.0% 0.0% 0.0% 0.0%
Moderate 2.7% 2.3% 0.8% 1.5% 1.1% 0.5%
Mild 9.3% 13.7% 11.0% 11.0% 12.2% 10.7%
None 88.0% 84.0% 88.2% 87.5% 86.6% 88.8%
Baseline Week 2 Week 12 Baseline Week 2 Week 12
Erythema
SB204
Vehicle
Severe 0.0% 0.0% 0.0% 0.0% 0.0% 0.0%
Moderate 10.4% 8.6% 4.8% 7.1% 4.0% 2.4%
Mild 19.0% 25.7% 18.5% 20.7% 19.8% 14.6%
None 70.5% 65.7% 76.7% 72.2% 76.2% 83.1%
Baseline Week 2 Week 12 Baseline Week 2 Week 12
Pruritus
SB204
Vehicle
Severe 0.0% 0.0% 0.0% 0.0% 0.0% 0.0%
Moderate 2.0% 1.2% 0.5% 2.2% 0.2% 0.2%
Mild 7.5% 5.6% 5.3% 5.6% 6.0% 2.9%
None 90.5% 93.3% 94.2% 92.2% 93.8% 96.9%
Baseline Week 2 Week 12 Baseline Week 2 Week 12
Scaling
SB204
Vehicle
Severe 0.0% 0.0% 0.0% 0.0% 0.0% 0.0%
Moderate 1.1% 1.6% 1.0% 0.6% 0.4% 0.2%
Mild 9.5% 10.6% 6.5% 8.0% 8.0% 6.0%
None 89.3% 87.7% 92.5% 91.4% 91.5% 93.5%
Baseline Week 2 Week 12 Baseline Week 2 Week 12

NI-AC301/302 Results: Efficacy

Inflammatory Lesions

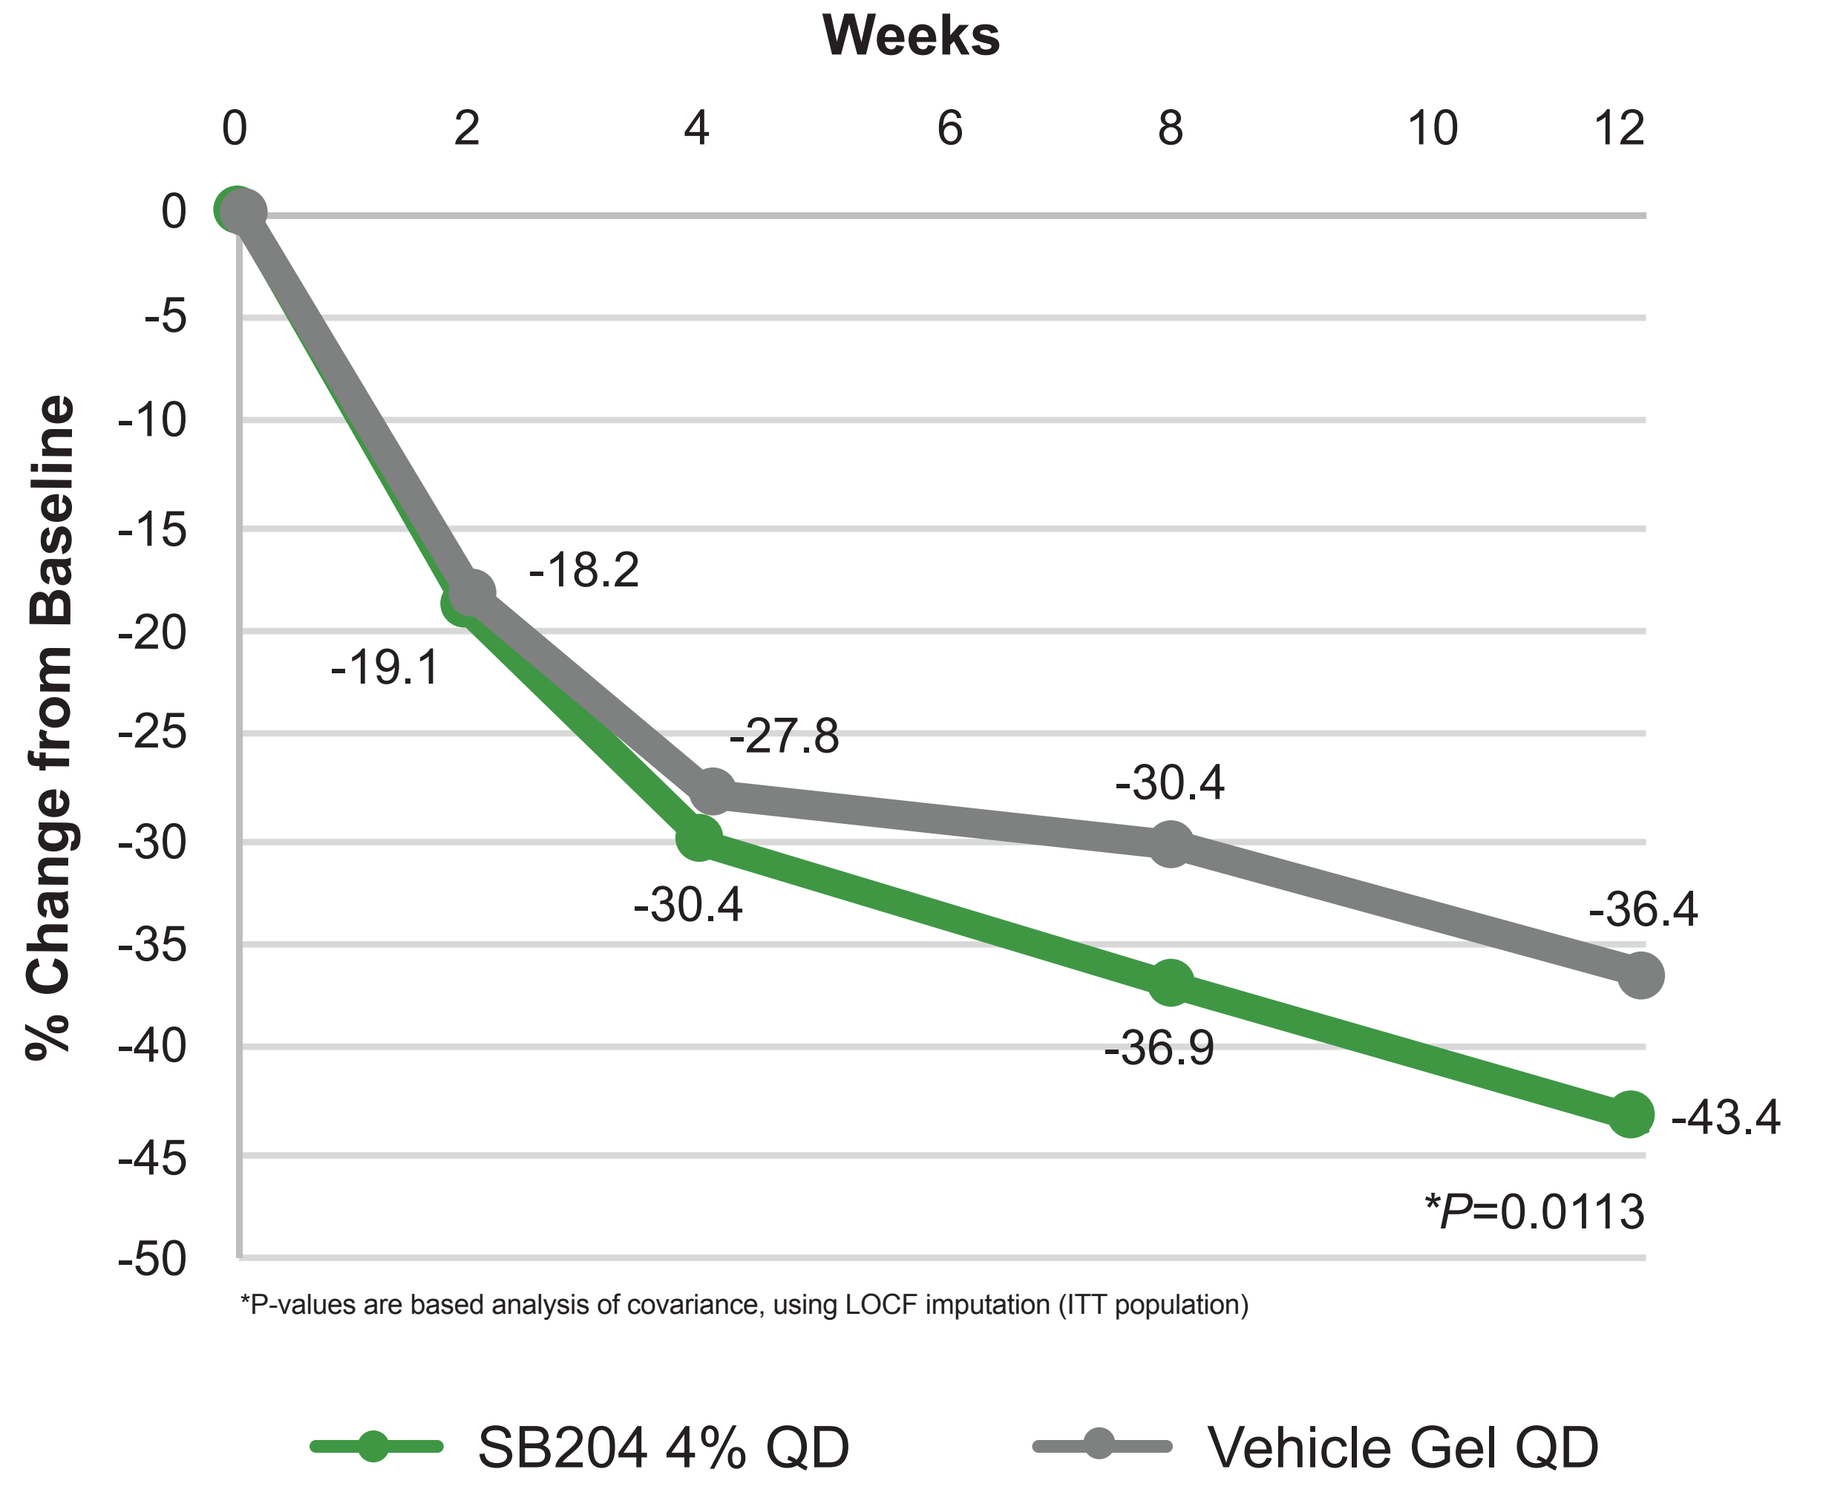
Weeks
0 2 4 6 8 10 12
0 -5 -10 -15 -20 -25 -30 -35 -40 -45 -50
% Change from Baseline
-19.1
-18.2
-27.8
-30.4
-30.4
-36.9
-36.4
-43.4
*P=0.0113
*P-values are based analysis of covariance, using LOCF imputation (ITT population)
SB204 4% QD
Vehicle Gel QD

Non-Inflammatory Lesions

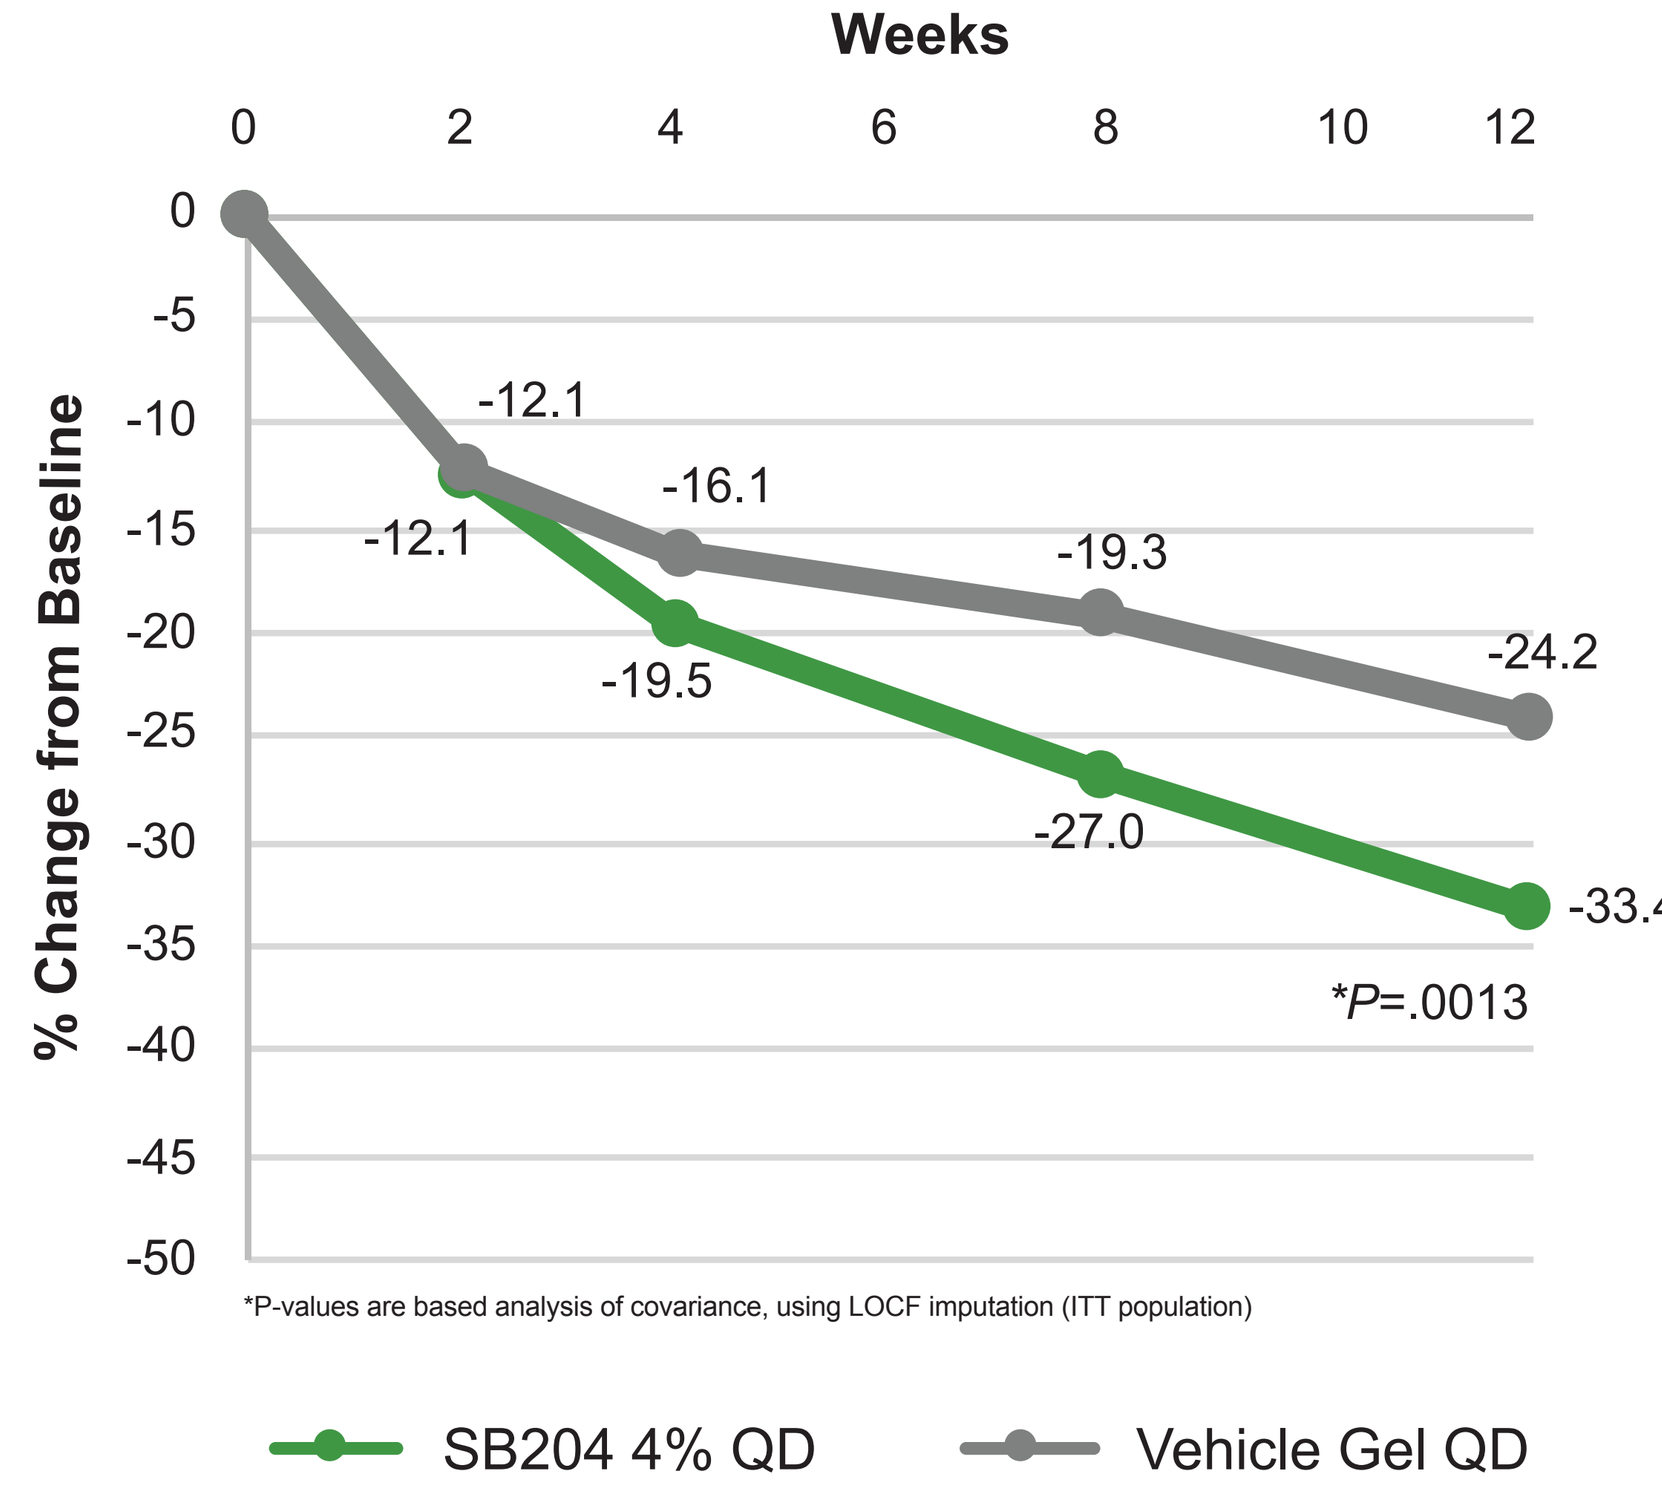
Weeks
0 2 4 6 8 10 12
0 -5 -10 -15 -20 -25 -30 -35 -40 -45 -50
% Change from Baseline
-12.1
-12.1
-16.1
-19.5
-19.3
-27.0
-24.2
-33.4
*P=.0013
*P-values are based analysis of covariance, using LOCF imputation (ITT population)
SB204 4% QD
Vehicle Gel QD

Total Lesions

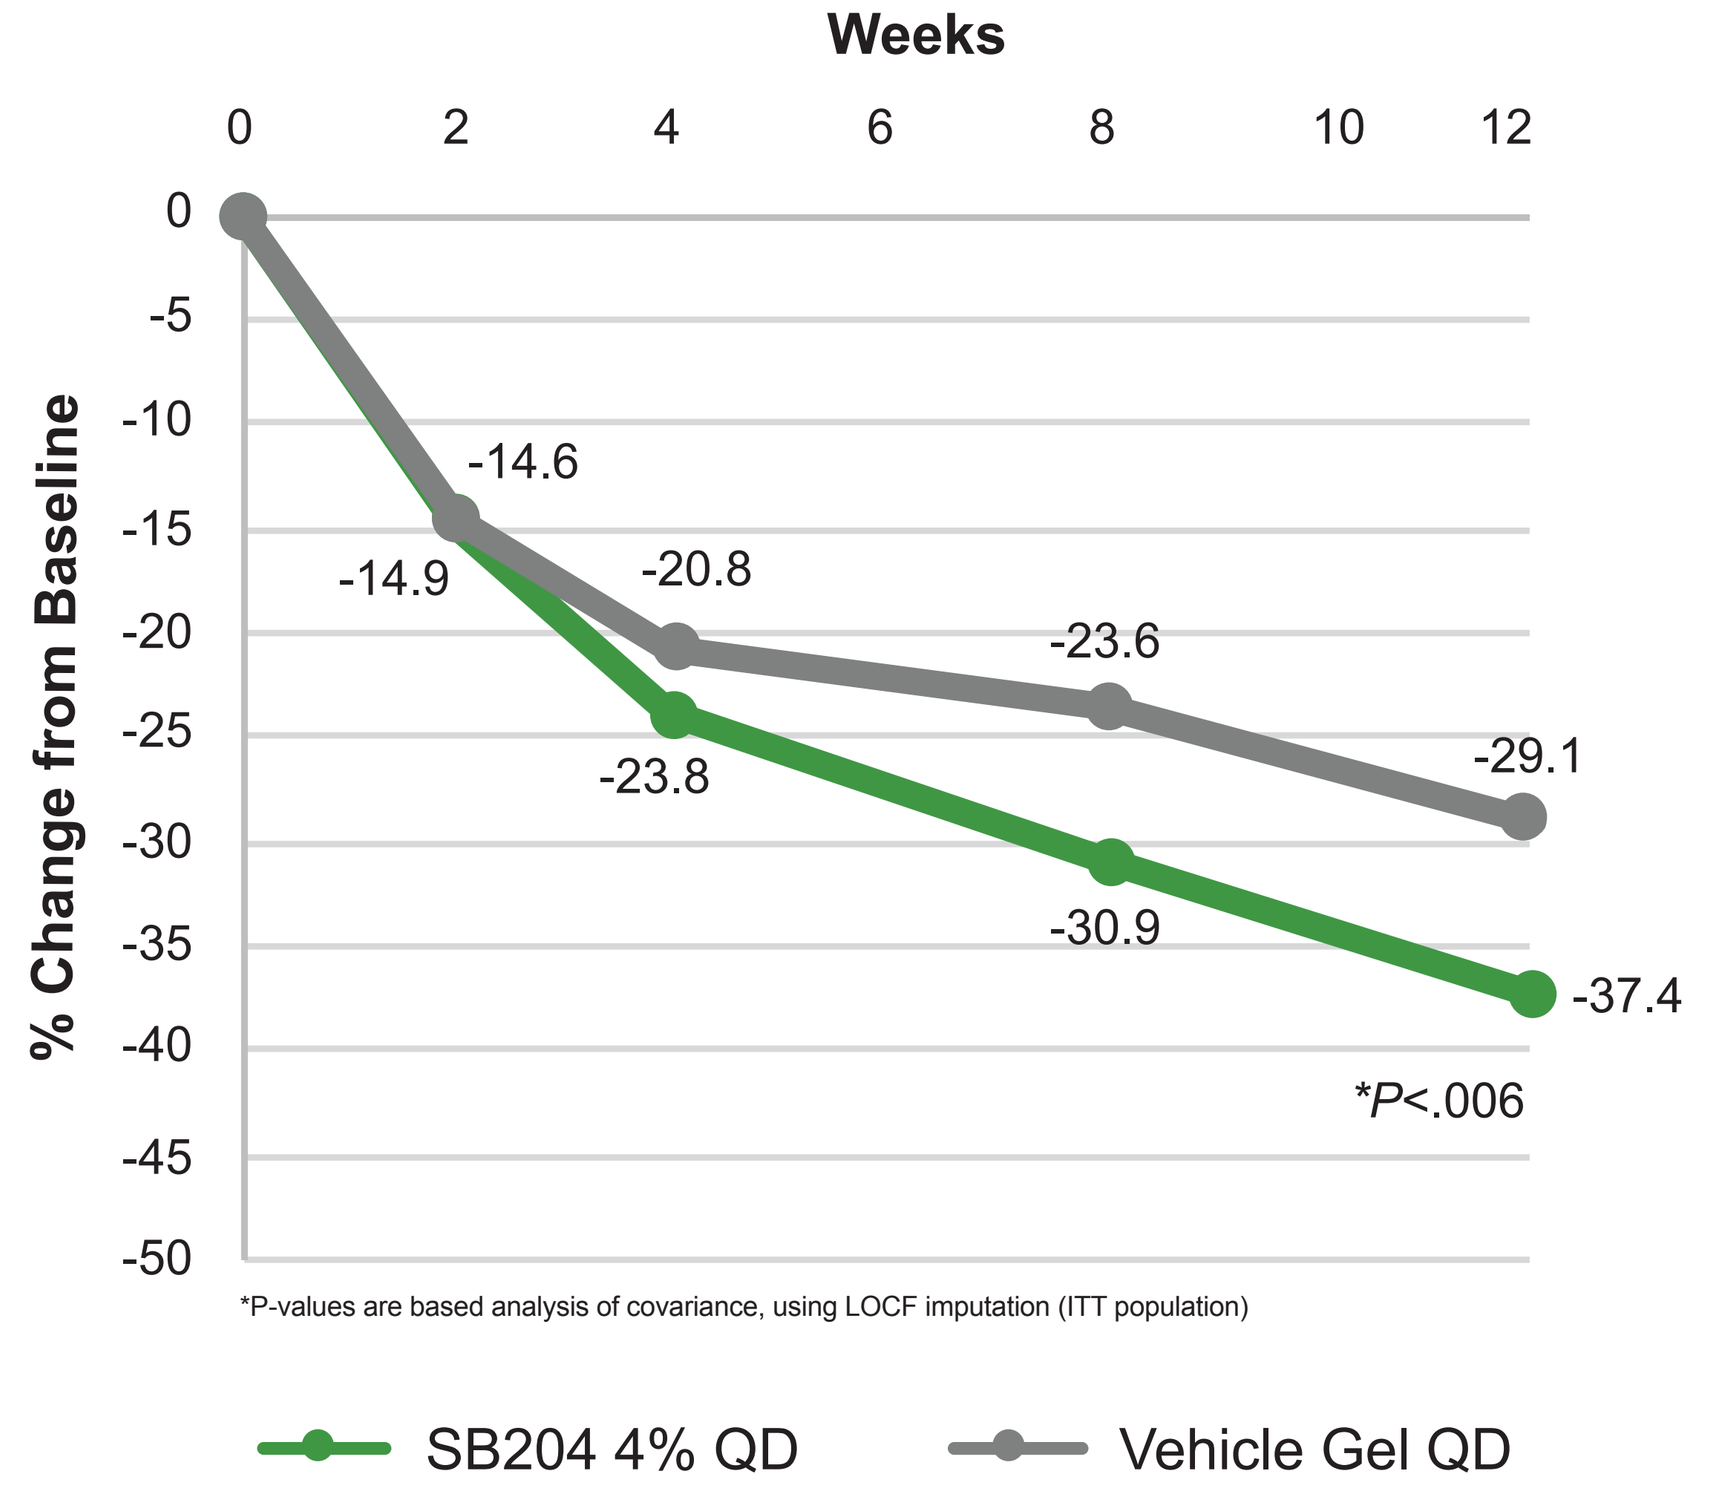
Weeks
0 2 4 6 8 10 12
0 -5 -10 -15 -20 -25 -30 -35 -40 -45 -50
% Change from Baseline
-14.6
-14.9
-20.8
-23.8
-23.6
-30.9
-29.1
-37.4
*P<.006
*P-values are based analysis of covariance, using LOCF imputation (ITT population)
SB204 4% QD
Vehicle Gel QD

NI-AC301/302 Treatment Emergent Adverse Events (TEAEs)

NI-AC301/302	Number of AEs	Dermatitis	Dryness	Erythema	Exfoliation	Pain	Pruritus	Rash	Reaction	Swelling	Malaise	Pyrexia
SB204 4% (N=439) n incidence	38	1 (0.23%)	3 (0.68%)	2 (0.46%)	1 (0.23%)	7 (1.59%)	3 (0.68%)	2 (0.46%)	1 (0.23%)	1 (0.23%)	1 (0.23%)	1 (0.23%)

NI-AC103 Study

NI-AC103 STUDY OVERVIEW

		NI-AC103 (N=18)
Study Design		Single-center, open-label pharmacokinetic study
Age		Age ≥9 - <17 yrs.
Dose/Dosing Schedule		Topical SB204 4% was applied once daily in the mornings for 21 days
PK Collection		PK profiling done on days 1 and 21 from time 0 to 24 hours post dose
Sex, N	Male	11 (61%)
	Female	7 (39%)
Completed, N		18 (100%)
Discontinued, N		0

- The following systemic exposure were assessed:
 - hydrolyzed N-methyl-aminopropyltrimethoxysilane (hMAP3), a component of the NVN1000 parent compound backbone. The lower limit of quantitation (LLOQ) for hMAP3 was 5.0 ng/mL.
 - Nitrate, a marker for systemic nitric oxide exposure. The LLOQ for nitrate was 300 ng/ml.
- Treatment Emergent Adverse Events (TEAEs) were recorded, methemoglobin was monitored and electrocardiogram (ECG) readings were taken

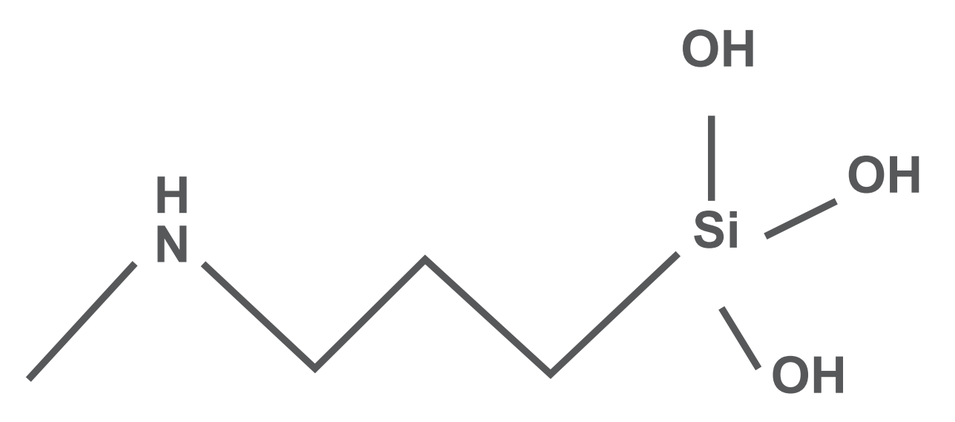
OH
OH
OH
Si
H
N

Hydrolyzed MAP3 (hMAP3)

NI-AC103 Treatment Emergent Adverse Events

- In all subjects, no clinically significant changes in laboratory assessments, including methemoglobin values, ECG results and physical examinations, were observed

NI-AC103	SB204 4% (N=18) n incidence
Number of subjects reporting at least one AE	1 (5.6%)
Number of AEs	2
Skin and subcutaneous system disorders	1 (5.6%)
Injury, poisoning, and procedural complications	1 (5.6%)

NI-AC103 Results

In the open label pharmacokinetic study (NI-AC103) in adolescents (age ≥9 - <17 yrs.; n=18):

- Plasma hMAP3 levels were below LLOQ (5.0 ng/mL) after single application SB204 4% at all timepoints
- There were no noticeable differences in nitrate PK parameters in subjects at baseline and following treatment with SB204 4%

Conclusions

- Following single and repeat application of topical SB204 4% in adolescent subjects with moderate to severe acne in NI-AC103:
 - There was no detectable hMAP3 in any subject, at all timepoints, and for all treatments studies, including under maximal use conditions
 - There were no statistically significant differences in nitrate PK parameters at baseline and following treatment with SB204
- SB204 4% **once daily** showed a statistically significant reduction in inflammatory, non-inflammatory and total lesions while maintaining a good safety and tolerability profile in adolescent subjects in NI-AC301 and NI-AC302
- All doses of SB204 administered in the studies were well tolerated and the adverse event profile was similar in active and vehicle treated subjects